Single-cell transcriptome analyses further elucidate the mechanism of action of EED inhibition on HbF induction

Summer Xia Han, Emily Fitz, Allysa Allen, Avik Choudhuri, Caroline V. Sartain, Paul A. Bruno, Jeffrey W. Jacobs, Billy Stuart

[1] Fulcrum Therapeutics, 26 Landsdowne Street, Cambridge, MA, USA

INTRODUCTION

Fetal Hemoglobin (HbF) Mitigates Mortality and Morbidity Risks Associated with Sickle Cell Disease

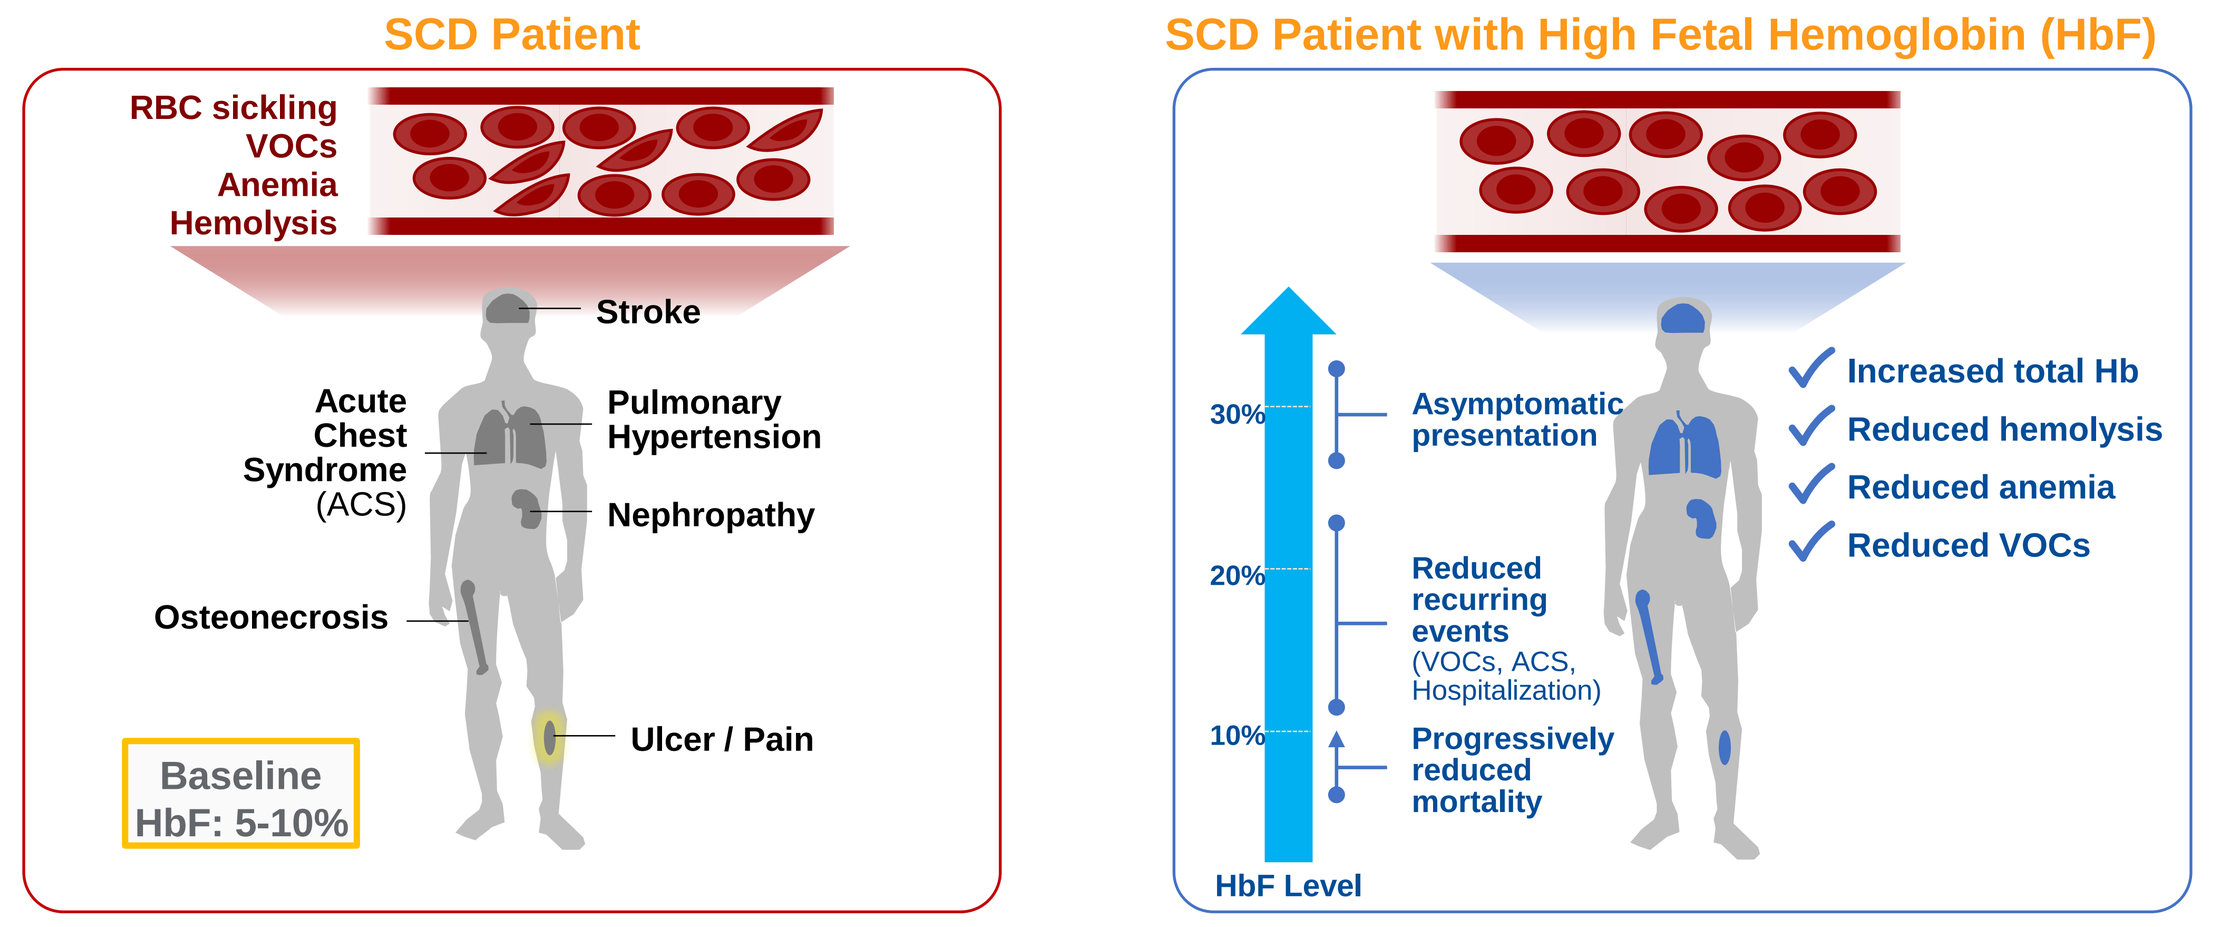
SCD Patient
RBC sickling
VOCs
Anemia
Hemolysis
Stroke
Acute Chest Syndrome (ACS)
Pulmonary Hypertension
Nephropathy
Osteonecrosis
Ulcer / Pain
Baseline HbF: 5-10%
SCD Patient with High Fetal Hemoglobin (HbF)
30%
20%
10%
Asymptomatic presentation
Reduced recurring events (VOCs, ACS, Hospitalization)
Progressively reduced mortality
HbF Level
Increased total Hb
Reduced hemolysis
Reduced anemia
Reduced VOCs

People living with SCD can have additional mutations that cause a condition known as hereditary persistence of fetal hemoglobin (HPFH), which leads to reduced or no symptoms in patients with SCD

Increased HbF levels over typical baseline measurements relieve the severity of symptoms like hemolysis, anemia, and VOCs in people living with Sickle Cell Disease

Screening Identified Embryonic Ectoderm Development (EED) as a Target for HbF Induction

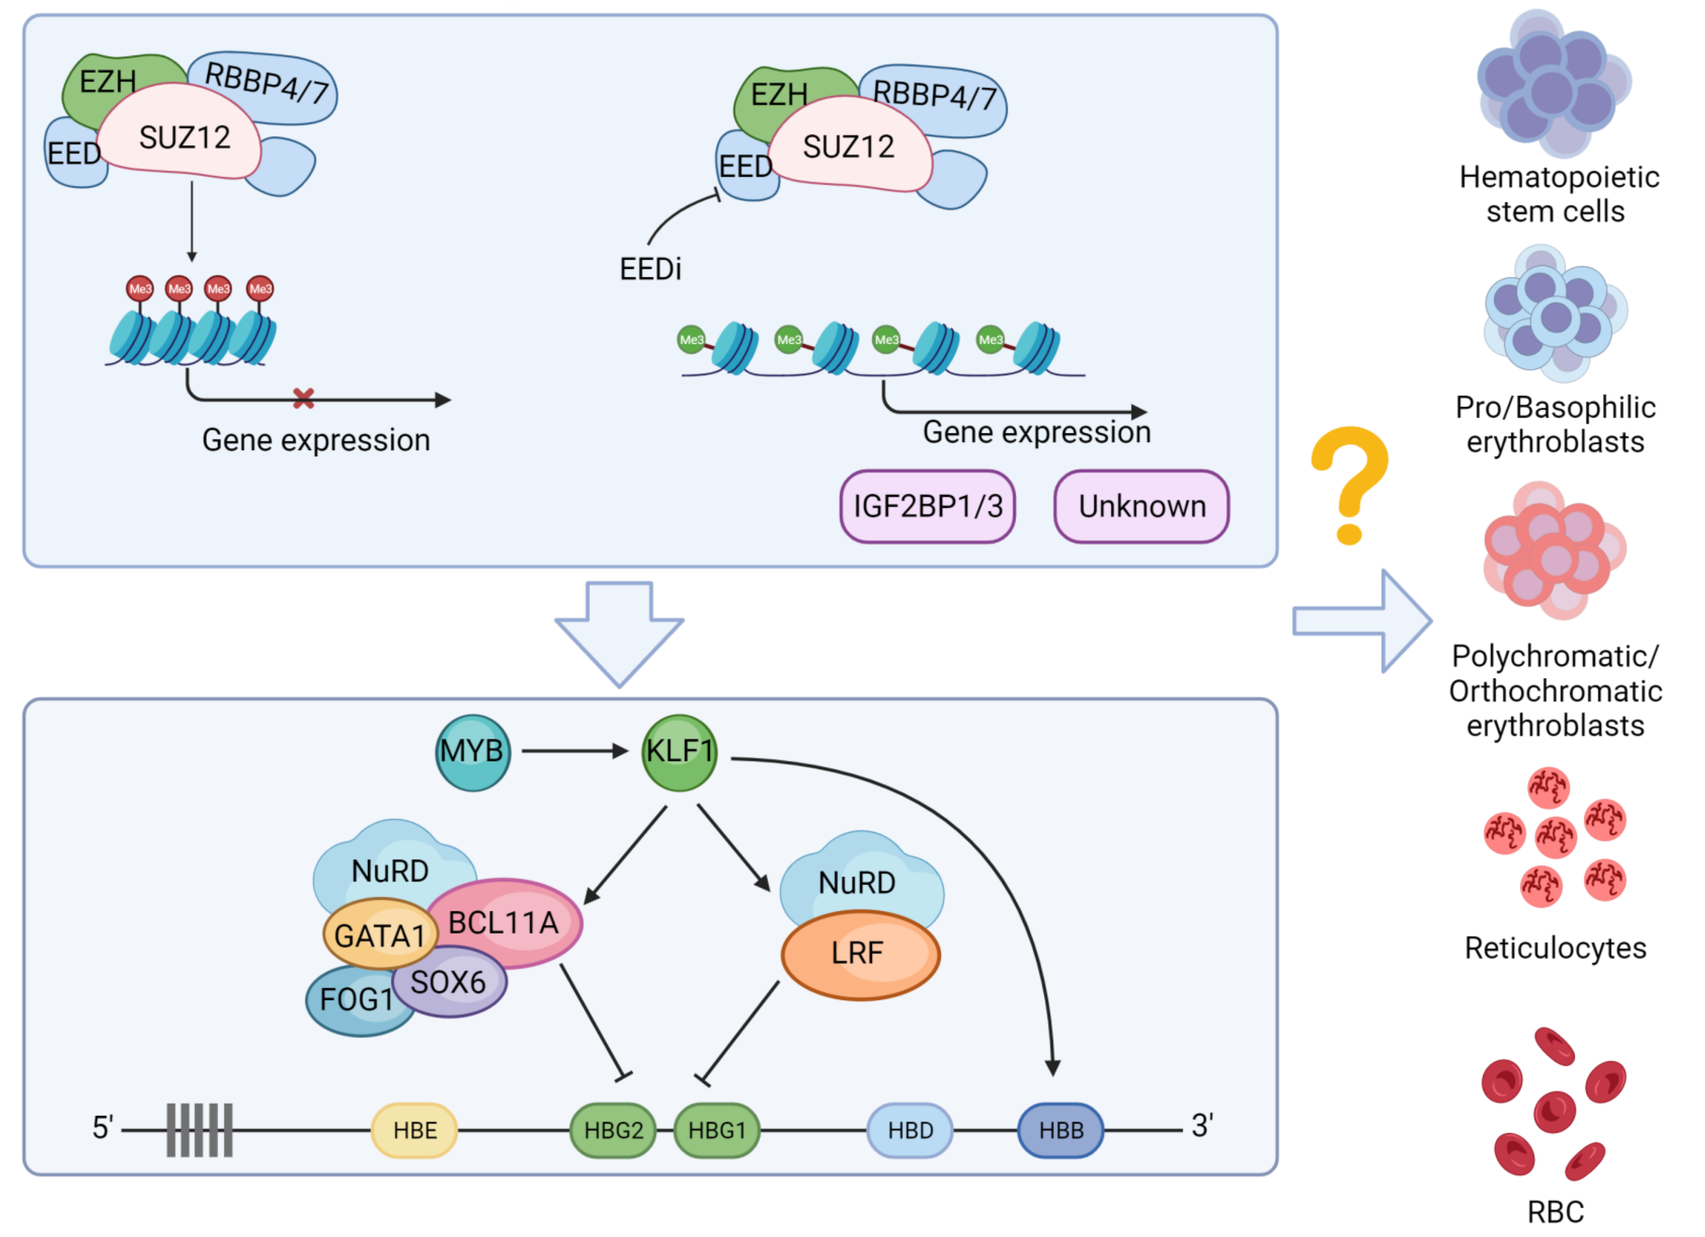
EZH
RBBP4/7
EED
SUZ12
Gene expression
EEDi
Gene expression
IGF2BP1/3
Unknown
MYB
KLF1
NuRD
GATA1
BCL11A
FOG1
SOX6
NuRD
LRF
5'
HBE
HBG2
HBG1
HBD
HBB
3'
Hematopoietic stem cells
Pro/Basophilic erythroblasts
Polychromatic/ Orthochromatic erythroblasts
Reticulocytes
RBC

EED regulates HbF expression through indirect interactions with the known HbF regulatory pathway. The clear molecular and cellular mechanism of actions of EED inhibitor (EEDi) remains to be investigated

RESULTS

A. Cell type identification of CD34+ hematopoietic stem cells (HSCs) derived erythroid culture

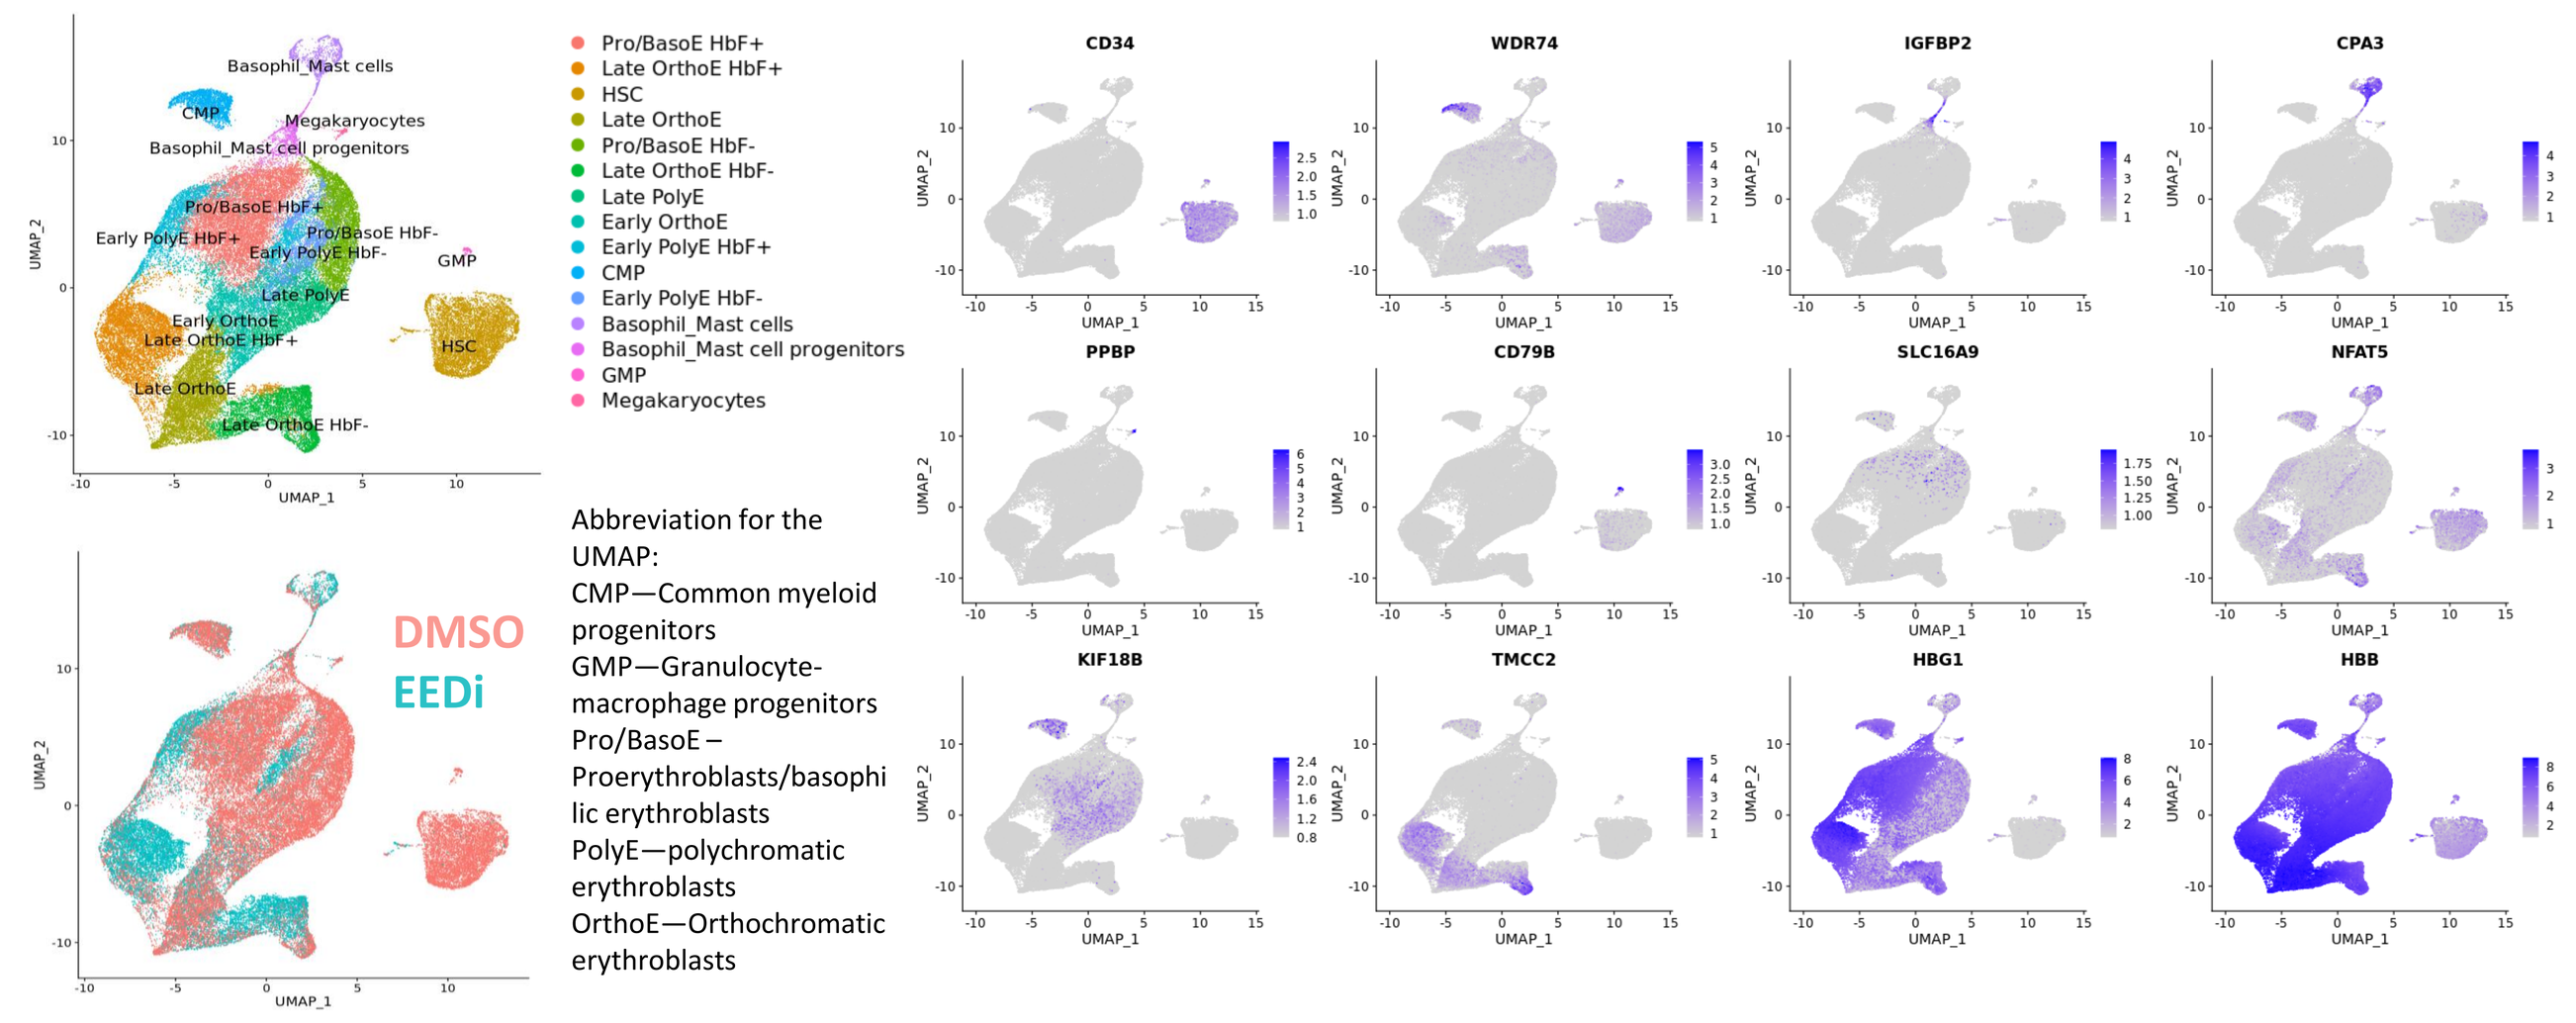
DMSO
EEDi
Abbreviation for the UMAP:
CMP—Common myeloid progenitors
GMP—Granulocyte-macrophage progenitors
Pro/BasoE – Proerythroblasts/basophilic erythroblasts
PolyE—polychromatic erythroblasts
OrthoE—Orthochromatic erythroblasts
CD34
WDR74
IGFBP2
CPA3
PPBP
CD79B
SLC16A9
NFAT5
KIF18B
TMCC2
HBG1
HBB

A) Left panel: Uniform Manifold Approximation and Projection (UMAP) representation of all the cells from different culture stages and treatment conditions show the presence of heterogeneous cell types derived from CD34+ HSCs. **Right panel:** Expression of cell type-specific markers overlaid on UMAP representation captures the presence from early progenitors to late erythroblasts. Values represent scaled count values for each gene.

B. Cell type compositions and pseudobulk expression of HbF-related genes change during differentiation

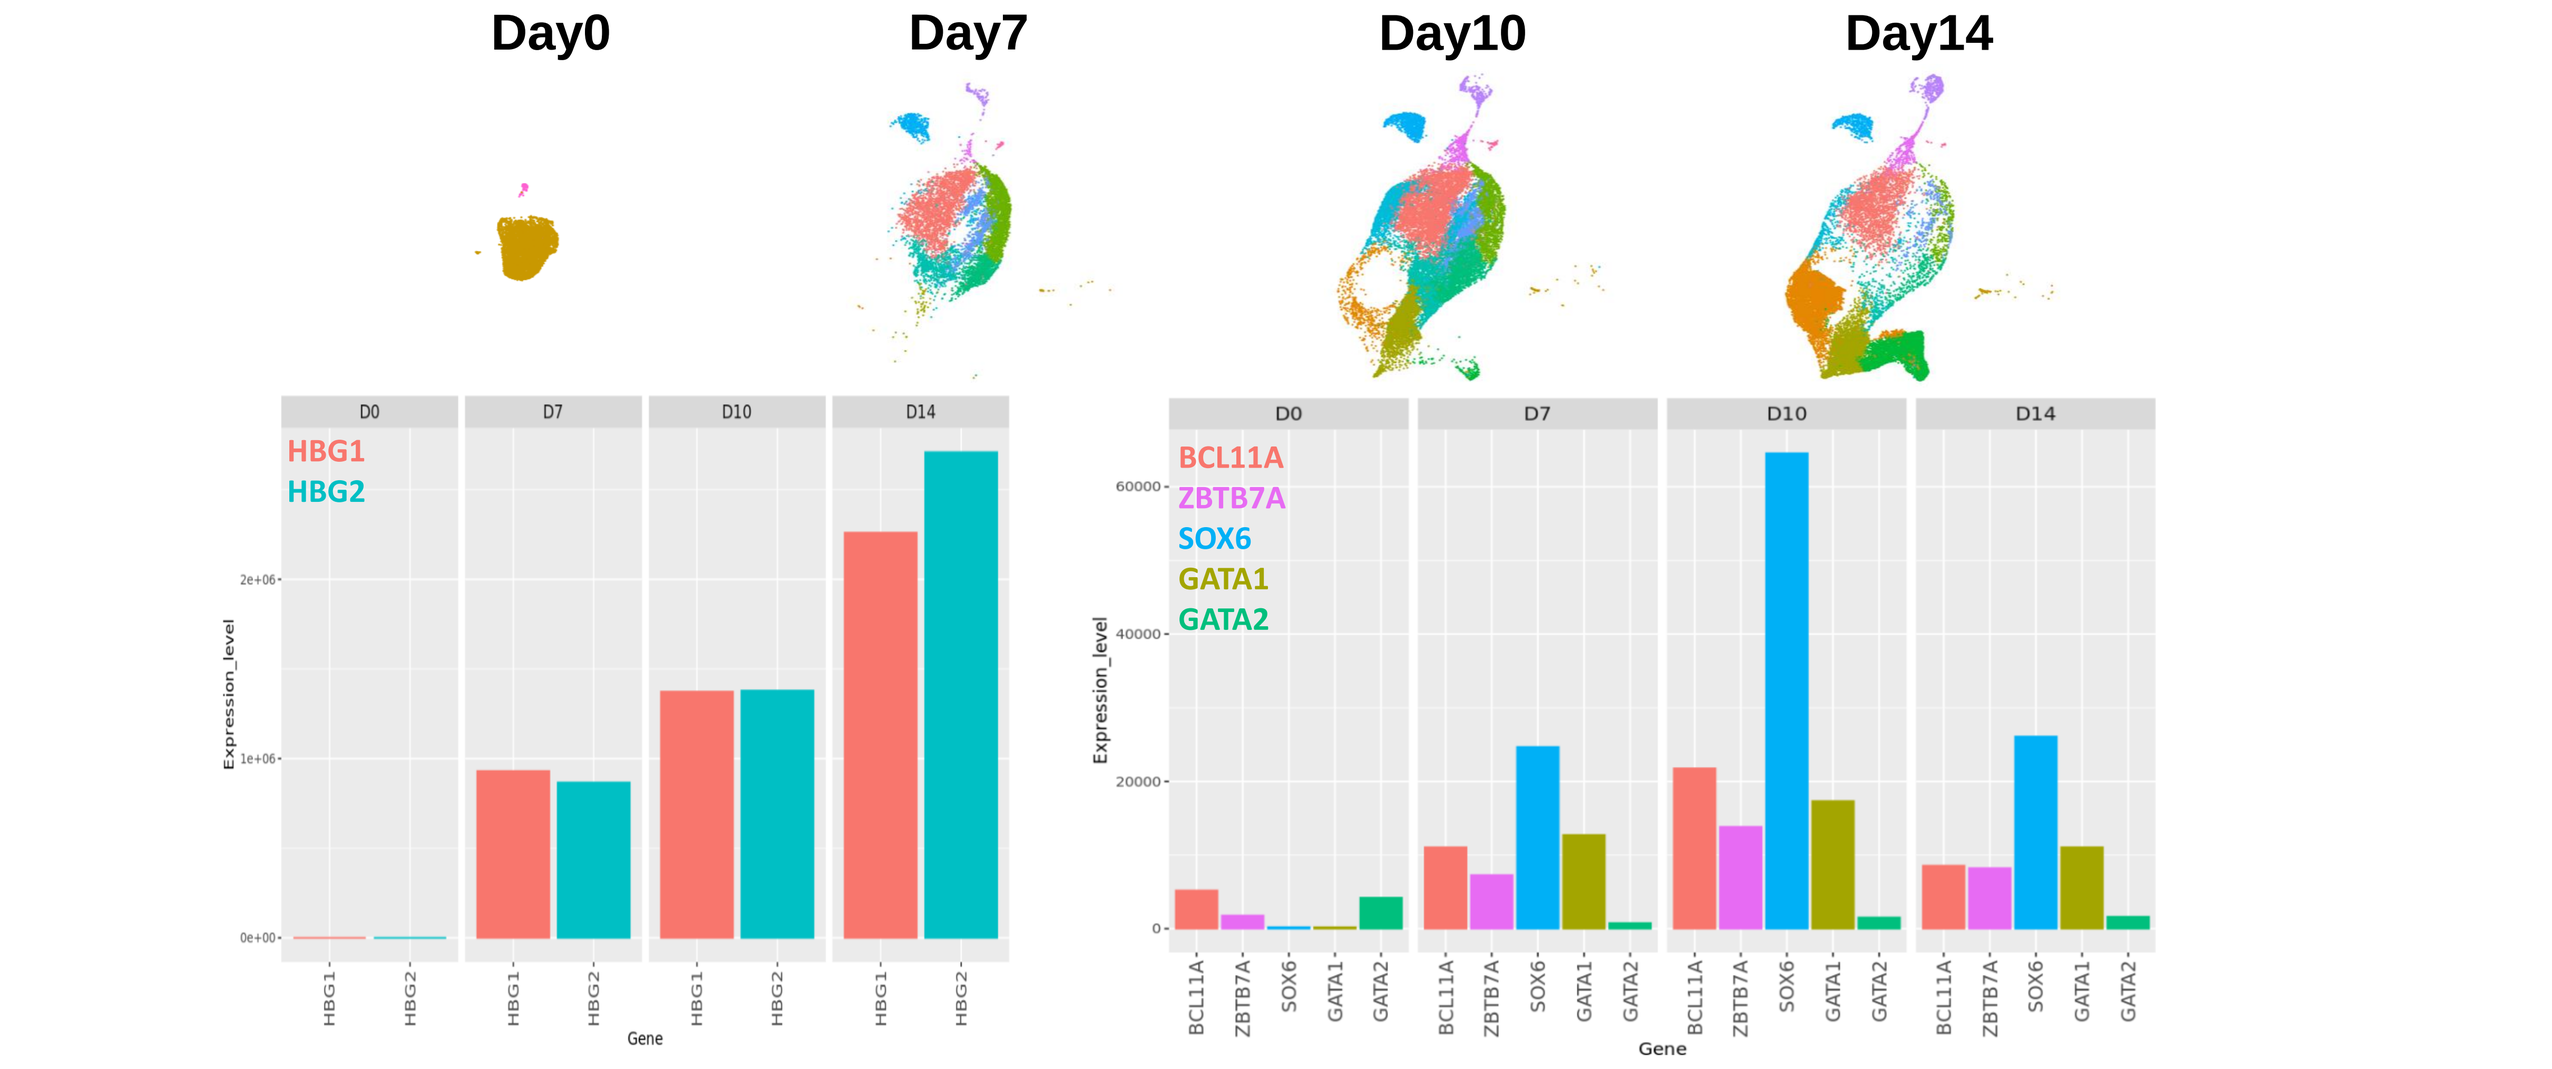
Day0
Day7
Day10
Day14
HBG1
HBG2
BCL11A
ZBTB7A
SOX6
GATA1
GATA2

B) UMAP representations show the majority of the cells start differentiating before Day 7. Comparison of cell compositions at different stages shows the differentiation direction of erythroblasts. Bar graphs of pseudobulk gene expressions at the lower panel verified the previously reported bulk expression patterns for HbF and their known regulators in our culture system.

C. Comparison of relative cell type composition between DMSO and EEDi

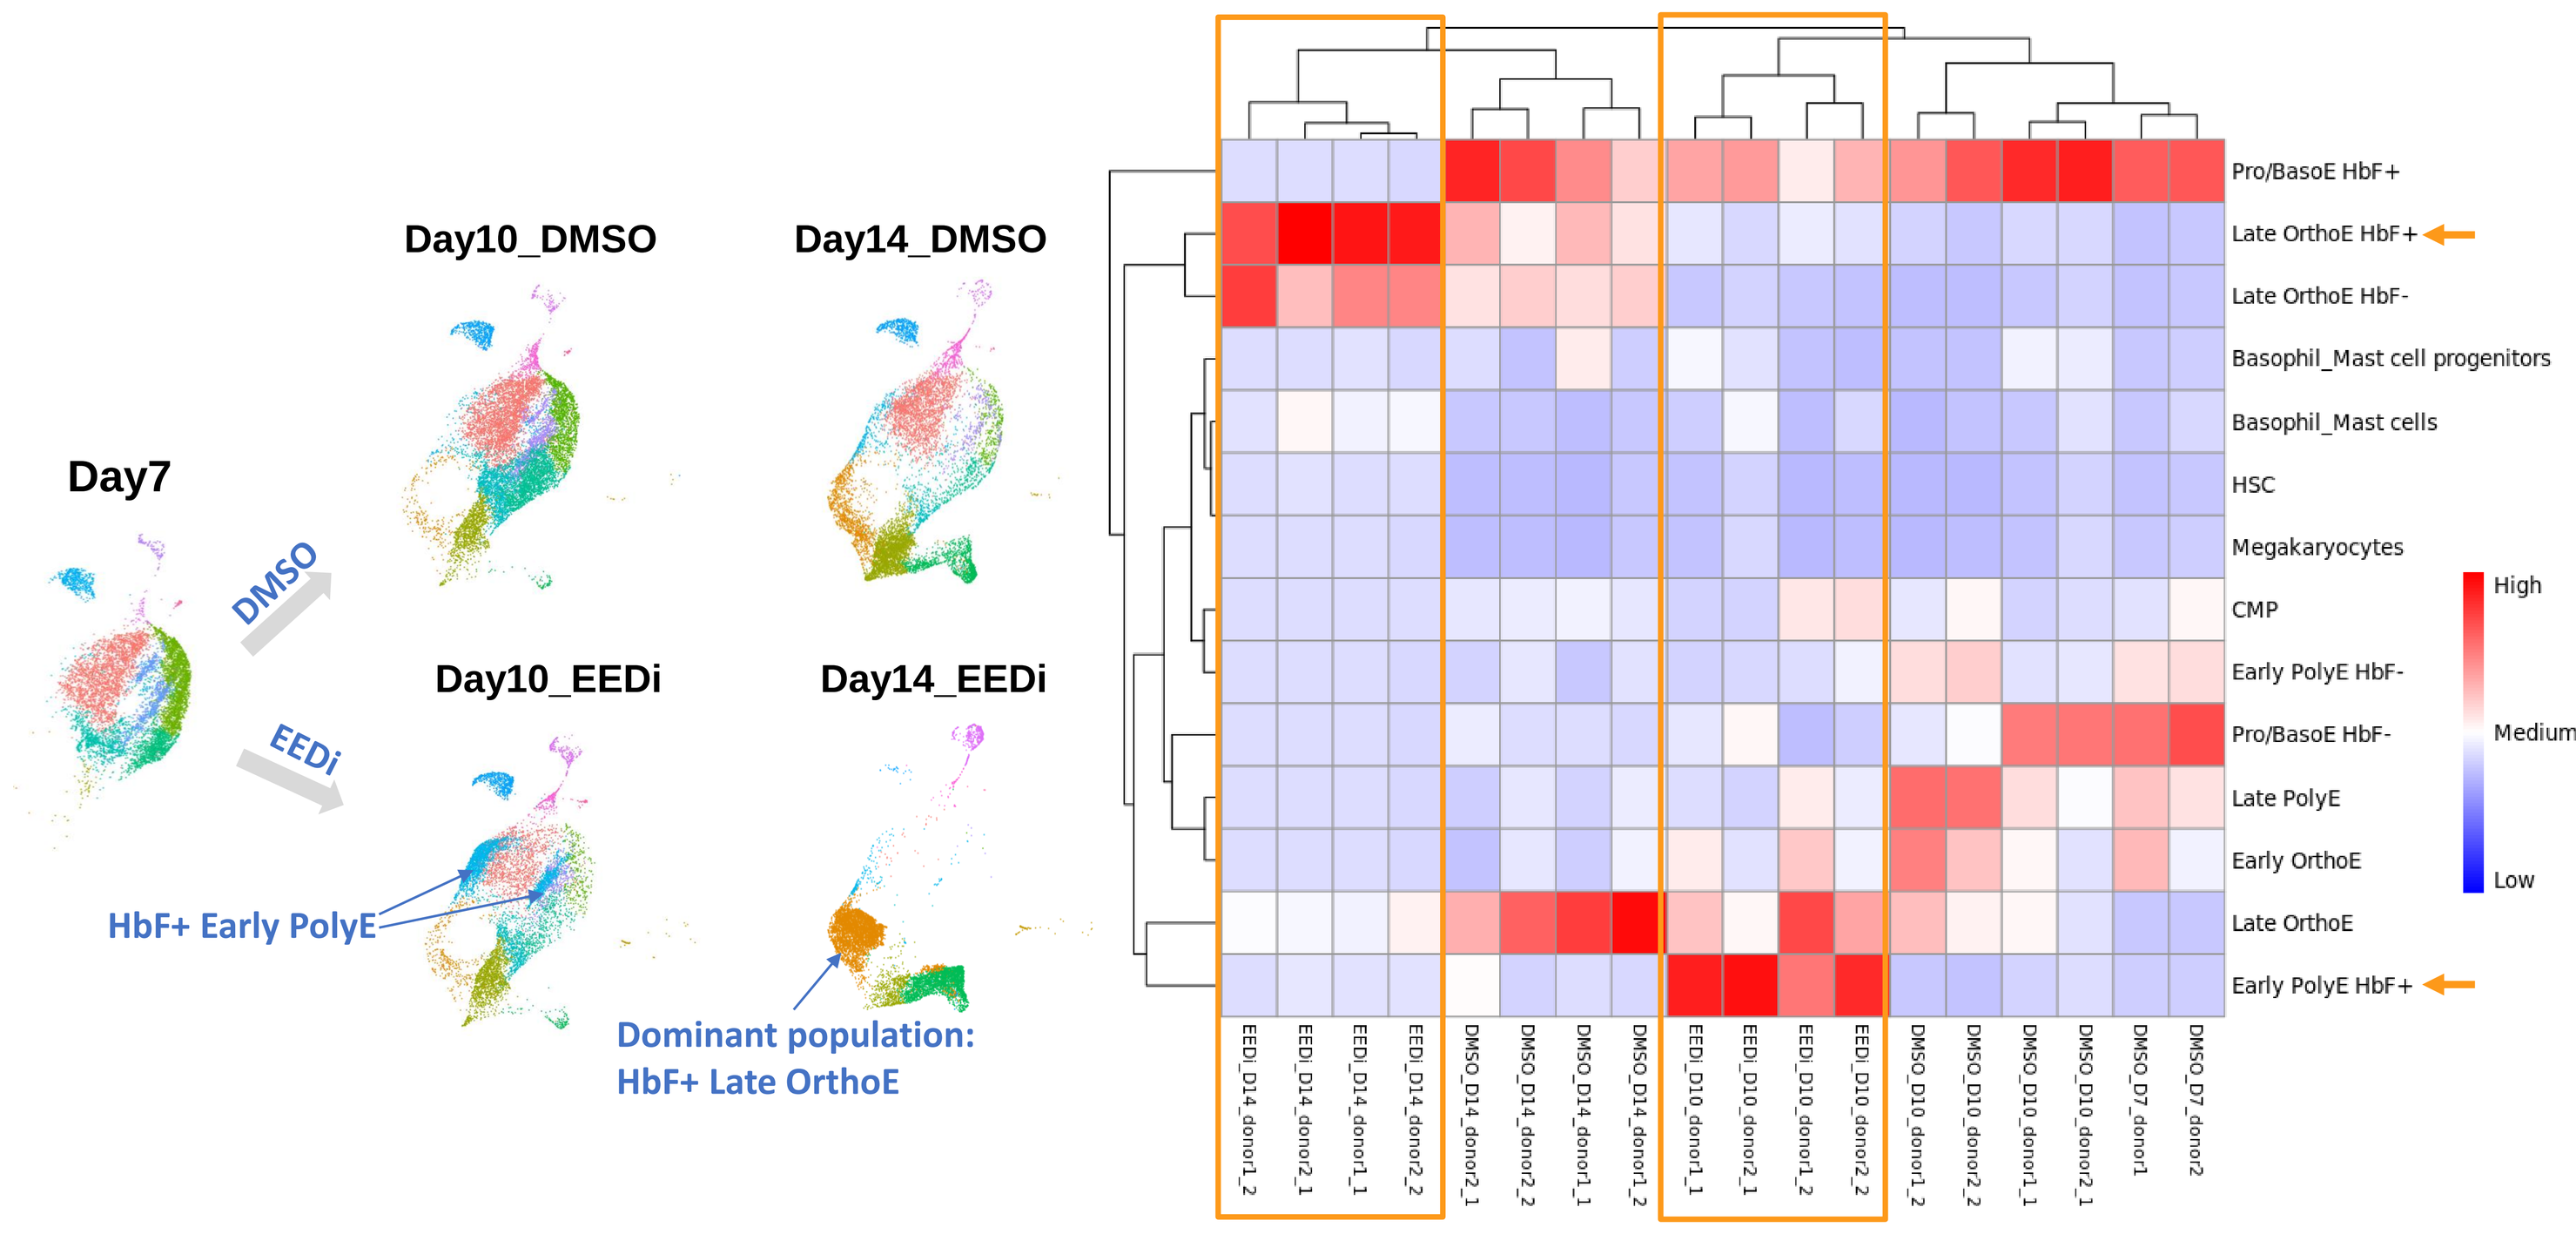
Day7
DMSO
EEDi
Day10_DMSO
Day14_DMSO
Day10_EEDi
Day14_EEDi
HbF+ Early PolyE
Dominant population: HbF+ Late OrthoE
Pro/BasoE HbF+
Late OrthoE HbF+
Late OrthoE HbF-
Basophil_Mast cell progenitors
Basophil_Mast cells
HSC
Megakaryocytes
CMP
Early PolyE HbF-
Pro/BasoE HbF-
Late PolyE
Early OrthoE
Late OrthoE
Early PolyE HbF+
High
Medium
Low

C) UMAP representation of cell populations under different treatment conditions show accelerated maturation and increase HbF+ population in response to EEDi treatment. The heatmap in the right panel suggests these cell composition changes are consistent between different donors and replicates.

D. Analysis of pseudobulk differentially expressed genes

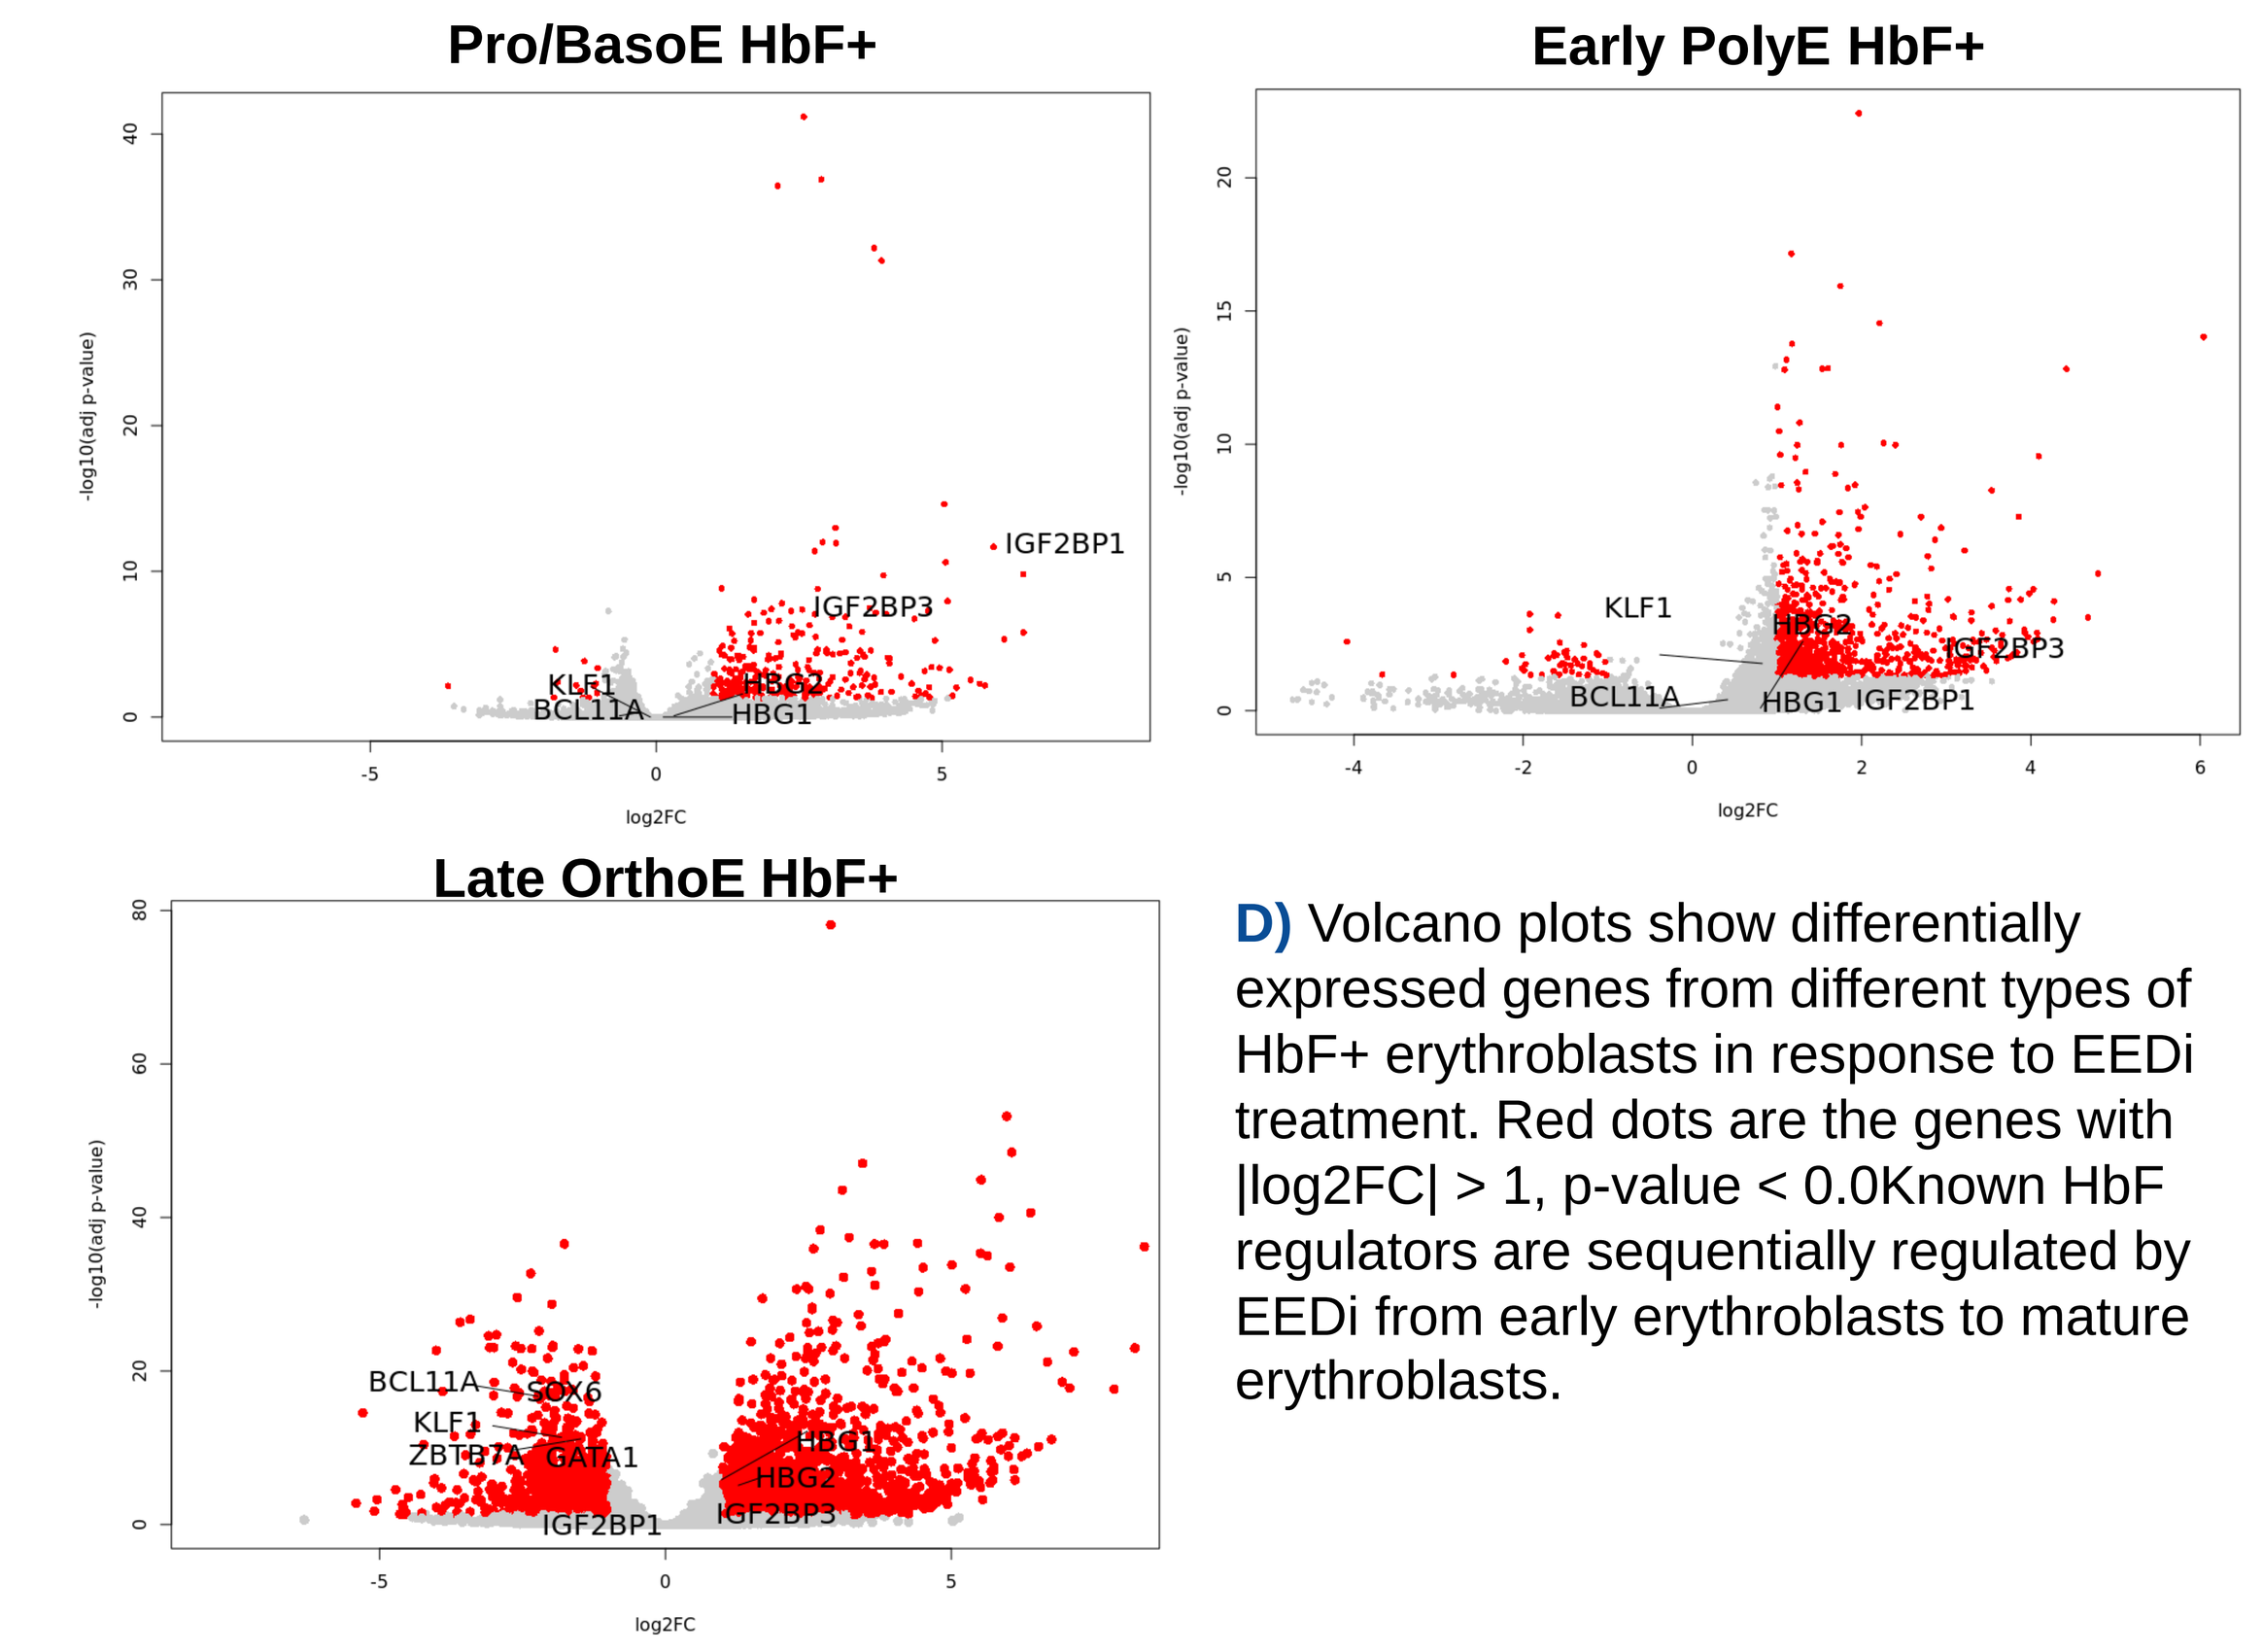
Pro/BasoE HbF+
Early PolyE HbF+
Late OrthoE HbF+
IGF2BP1
IGF2BP3
KLF1
BCL11A
HBG2
HBG1
SOX6
ZBTB7A
GATA1

D) Volcano plots show differentially expressed genes from different types of HbF+ erythroblasts in response to EEDi treatment. Red dots are the genes with |log2FC| > 1, p-value < 0.0Known HbF regulators are sequentially regulated by EEDi from early erythroblasts to mature erythroblasts.

E. Pseudotime analyses of DMSO vs EEDi treated CD34+ HSCs derived erythroid culture

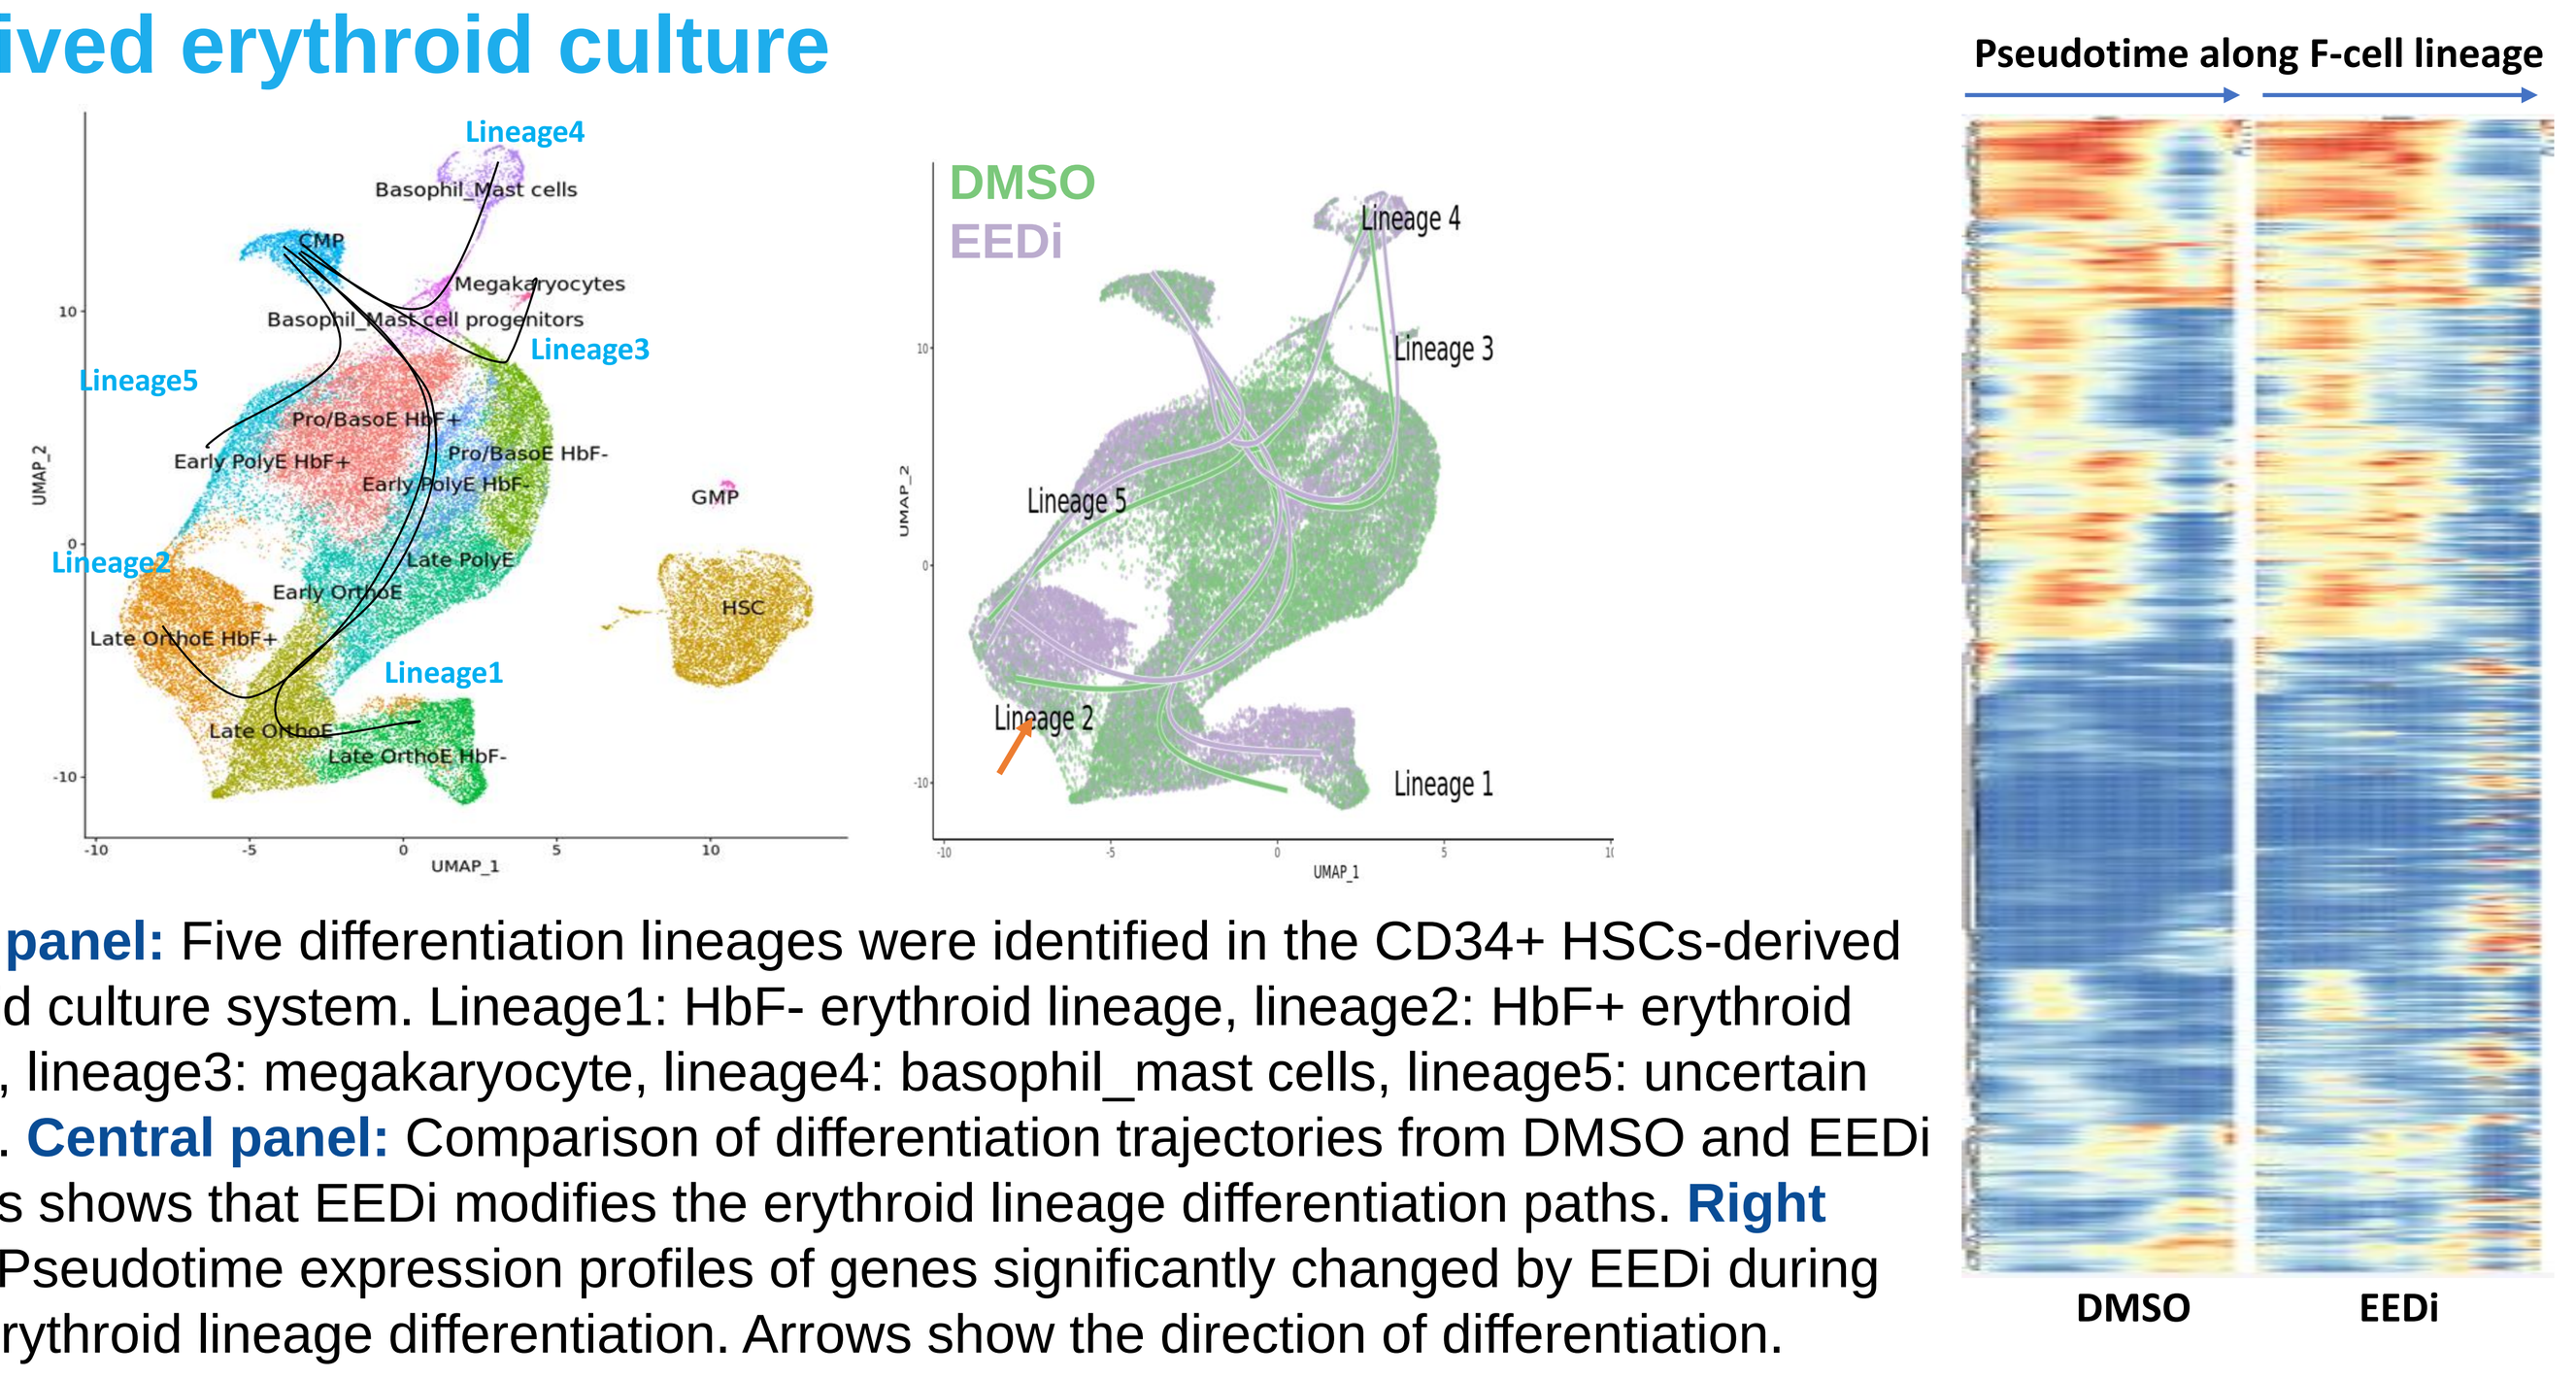
Lineage4
Lineage5
Lineage3
Lineage2
Lineage1
DMSO
EEDi
Lineage 4
Lineage 3
Lineage 5
Lineage 2
Lineage 1
Pseudotime along F-cell lineage
DMSO
EEDi

E) Left panel: Five differentiation lineages were identified in the CD34+ HSCs-derived erythroid culture system. Lineage1: HbF- erythroid lineage, lineage2: HbF+ erythroid lineage, lineage3: megakaryocyte, lineage4: basophil_mast cells, lineage5: uncertain lineage. **Central panel:** Comparison of differentiation trajectories from DMSO and EEDi samples shows that EEDi modifies the erythroid lineage differentiation paths. **Right panel:** Pseudotime expression profiles of genes significantly changed by EEDi during HbF+ erythroid lineage differentiation. Arrows show the direction of differentiation.

F. Pseudotime profiles of HbF-related genes

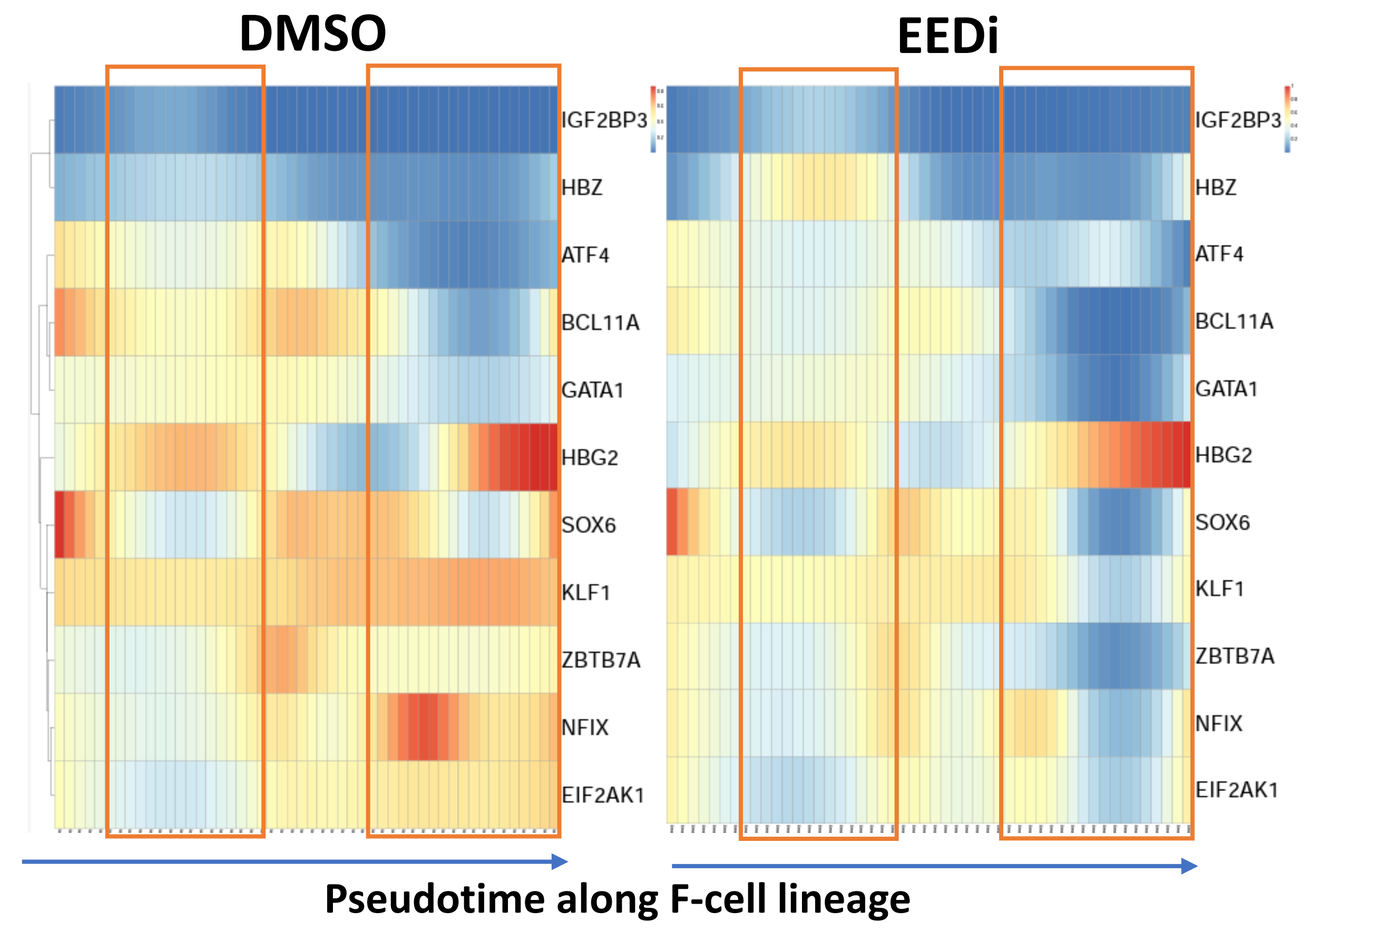
DMSO
EEDi
IGF2BP3
HBZ
ATF4
BCL11A
GATA1
HBG2
SOX6
KLF1
ZBTB7A
NFIX
EIF2AK1
Pseudotime along F-cell lineage

F) Different types of HbF regulators are changed at different time points during HbF+ erythroid lineage differentiation. Arrows show the direction of differentiation.

G. Number of genes affected by EEDi along different lineages

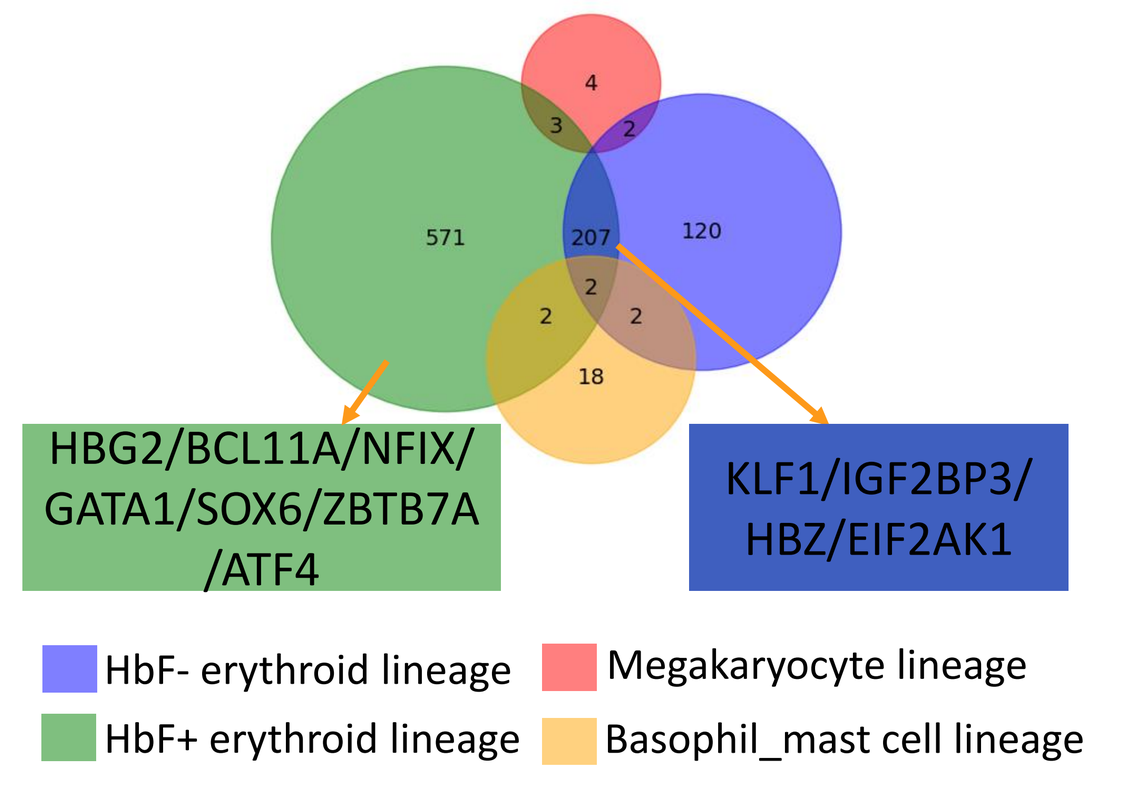
HBG2/BCL11A/NFIX/ GATA1/SOX6/ZBTB7A /ATF4
KLF1/IGF2BP3/ HBZ/EIF2AK1
HbF- erythroid lineage
Megakaryocyte lineage
HbF+ erythroid lineage
Basophil_mast cell lineage

G) Downstream target genes of EEDi show lineage specificity. Genes that are specifically affected in the HbF+ lineage are highlighted in the green box. Genes that are affected in both HbF- and HbF+ lineages are highlighted in the blue box.

METHODS

CD34+ hematopoiesis stem cells (HSC) derived erythroid culture:
Mobilized peripheral CD34+cells from two different donors were thawed in expansion media, comprised of IMDM basal medium, 100ng/mL hSCF, 5 ng/mL IL-3, 3 IU/mL EPO, 250 ug/mL Transferrin, 2.5% Normal Human Serum AB, 1% Penicillin-Streptomycin, 10 ng/mL Heparin and 10 ug/mL Insulin. After 7 days of expansion, cells were seeded into fresh expansion media, and compound treatment was applied (100nM EEDi or DMSO). On day 10, the media was exchanged to differentiation media which contains all expansion media components except for IL-3, and compound treatment was reapplied. The cells were differentiated until day 14 when they were harvested.

Sample collection and single-cell Isolation:
Cells were collected on day 0, day 7, day 10, and day 14. Cell viability was first measured by Vi-Cell blu cell viability analyzer (Beckman Coulter). For samples with >85% viability, individual cells were collected by filtering suspended culture through a 40-um cell strainer (Sigma-Aldrich). For samples with <85% viability, cells were stained with the viability dye Zombie NIR (Biolegend # 423106) and nuclei dye Hoechst33342 (Invitrogen H3570). After staining, cells were washed three times before loading to a Sony SH800 flow sorter. Viable single cells sorted and resuspended according to the 10X Genomics sample preparation protocol.

Library Preparation and Sequencing:
5000 cells from each sample were loaded into the 10X Chromium system and prepared for single-cell library construction using the 10X Genomics Chromium single cell 3' v3 reagent kit.

Library quality was analyzed by bioAnalyzer (Agilent). Libraries were quantified by the KAPA library quantification kit (Roche 07960336001). Sequencing was performed on the Novaseq 6000 platform with 150bp paired-end sequencing (Illumina).

Data analysis:
Library quality control, sequence alignment, and read counts were analyzed using the CellRanger pipeline version 7.0.1. Raw read counts from each single cell in each sample were analyzed using the Seurat 4.0 R package (Stuart et al., 2019). Cells with low reads and high mitochondrial ratios were filtered out, and each sample was normalized by SCTransform. To compare samples side by side, we performed integration analysis by combining the single-cell data from all the experimental conditions. Shared variances between different datasets were identified using the function SelectIntegrationFeatures and FindIntegrationAnchors based on canonical correlation analysis, then Seurat objects were processed using IntegrateData and SCTransform function. RunPCA and RunUMAP were then used for dimension reduction. To determine the resolution for cluster identification, multiple resolutions were tested by clustree analysis. Resolution 0.5 was chosen to find clusters.

Differentially expressed genes from each cell type were identified by pseudobulk analysis using DEseq from the Libra R package (Squair et al., 2021). Pseudotime trajectory analysis was performed by Slingshot(Street et al., 2018) under default setting to show cell differentiation paths across different culture time points. Using the Kolmogorov-Smirnov test, we assessed whether differences existed between the pseudotime distributions of the DMSO and EEDi treatment conditions. Subsequently, genes with different expression patterns along the trajectory between the two conditions were identified by tradeSeq (v1.6.0) (Van der Berge et al., 2020). For each condition, a negative binomial generalized additive model (fitGAM, nknots = 7) was applied to estimate a smoothed expression profile along the inferred trajectory for each gene. The fitted model was used for the conditionTest function to test whether genes exhibited different expression patterns along the differentiation paths between DMSO and EEDi treatment. Genes with adjusted p-values < 0.05 were identified based on the Wald test.

CONCLUSIONS

- We report first-in-class single-cell profiling of a CD34+ HSCs-derived erythroid culture system and show the heterogenous cell populations that form during erythroid differentiation.
- Comparison of cell populations of samples treated with and without EEDi reveals that EEDi treatment consistently leads to increased HbF+ populations and accelerated maturation across multiple donors without impairing erythroid cell maturation.
- Further analyses reveal that HbF+ and HbF- maturing erythroid cells undergo fate bifurcation. Specifically, the HbF+ erythroid differentiation path from EEDi-treated cell populations deviates from the differentiation path that control cells take starting from the late OrthoE population.
- Differential expression analysis along the HbF+ lineage identified multiple known HbF regulators as downstream targets of EEDi. Different types of downstream targets exhibit sequential changes in response to EEDi during differentiation.
- EEDi treatment leads to an erythroid-specific response, especially in the HbF+ erythroid lineage.

REFERENCES

Powars, DR et al. Is there a threshold level of fetal hemoglobin that ameliorates morbidity in sickle cell anemia? Blood (1984) Apr; 63(4):921-6

Platt, OS et al. Mortality in sickle cell disease. Life expectancy and risk factors for early death. NEJM (1994); 9;330(23):1639-44.

Akinsheye, I et al. Fetal hemoglobin in sickle cell anemia. Blood (2011) Jul 7; 118(1):19-27.

Stuart, T *et al.* Comprehensive integration of Single-Cell data. Cell (2019) 177:1888–1902.

Squair, JW *et al.* Confronting false discoveries in single-cell differential expression. *Nat Commun.* (2021)**12**, 5692.

Street, K *et al.* Slingshot: cell lineage and pseudotime inference for single-cell transcriptomics. *BMC Genomics* (2018)**19**, 477.

Van den Berge *et al.* Trajectory-based differential expression analysis for single-cell sequencing data. *Nat Commun* (2020) **11**, 1201.

CONTACT INFORMATION

Please contact us at info@fulcrumtx.com

www.fulcrumtx.com